#1964

Firmonertinib (furmonertinib) is a highly brain-penetrant EGFR inhibitor broadly active against uncommon EGFR mutations, including PACC and exon 20 insertions

Monique B. Nilsson[1], Zineb Mounir[2], Luna Musib[2], Junqin He[1], Xiaoxing Yu[1], Qing Li[3], Huibing Luo[3], John V. Heymach[1], Jerry Y. Hsu[2], Stuart Lutzker[2], Xiuning Le[1], Marcin Kowanetz[2]

Background

- Firmonertinib (rINN; also known as furmonertinib) is an oral, highly brain-penetrant, and broadly active mutation-selective epidermal growth factor receptor (EGFR) inhibitor active against both classical and uncommon EGFR mutations (Musib et al, 2022).
- It is approved in China for first-line advanced non-small-cell lung cancer (NSCLC) with EGFR exon 19 deletion or L858R mutations based on progression-free survival (PFS) benefit observed in the Phase 3 study vs gefitinib (FURLONG), as well as for patients with previously treated locally advanced or metastatic NSCLC with EGFR T790M mutation.
- It is currently being studied in a global Phase 3 trial for first-line NSCLC patients with EGFR exon 20 insertion mutations (FURVENT; NCT05607550), as well as in a global Phase Ib study evaluating firmonertinib in patients with activating EGFR or HER2 mutations, including uncommon EGFR mutations (FURTHER; NCT05364043).
- Firmonertinib has been granted US FDA Breakthrough Therapy Designation for the treatment of patients with previously untreated locally advanced or metastatic nonsquamous NSCLC with EGFR exon 20 insertion mutations.
- EGFR exon 20 insertion mutations are uncommon EGFR mutations that activate EGFR and result in the addition of 1 or more amino acids, typically clustered between positions E762 and C775 within the EGFR kinase domain, which alters the drug binding pocket to impede binding by drugs such as osimertinib; more than 120 variants have been identified (Friedlaender et al, 2022).
- P-loop αC-helix compressing (PACC) mutations represent another subset of uncommon EGFR activating mutations (Robichaux et al, 2021) that are similar to exon 20 insertion mutations in altering the drug binding pocket and are found as either single or compound EGFR mutations.
- Here, we further characterized PACC mutations as cancer drivers in EGFR-mutated NSCLC and evaluated the preclinical activity and binding mechanisms of firmonertinib against mutant EGFR with PACC and exon 20 insertion mutations.

Figure 1. PACC and exon 20 insertion mutations represent ~22% of EGFR mutations in NSCLC

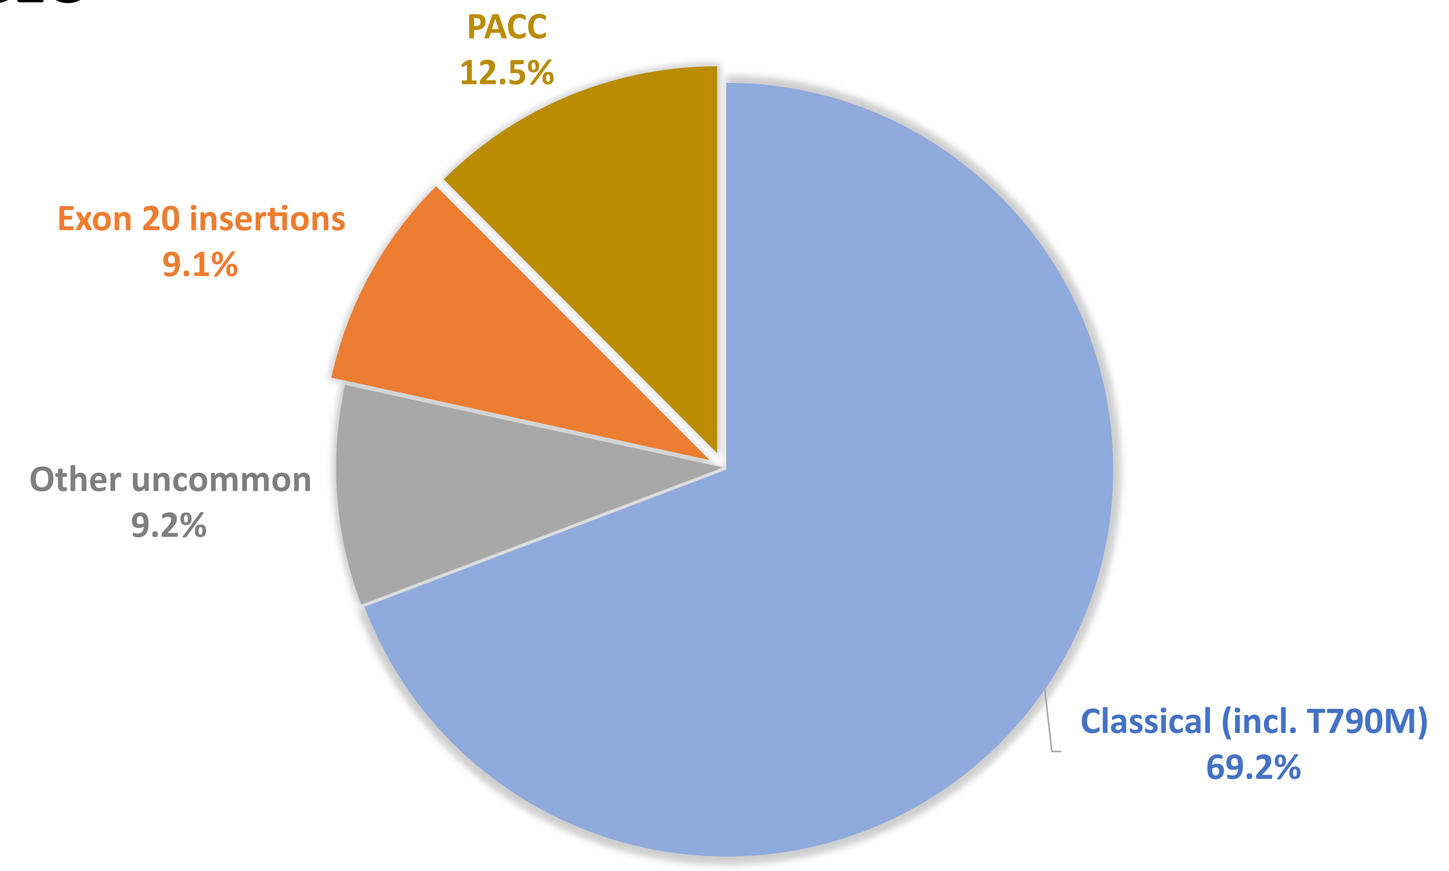
PACC
12.5%
Exon 20 insertions
9.1%
Other uncommon
9.2%
Classical (incl. T790M)
69.2%

Pie chart representing frequency of various EGFR mutations among all EGFR mutations in NSCLC. Modified from Robichaux et al, 2021. Percentage of PACC was calculated based on frequency of uncommon mutations in exon 18, 19, and 20 (after excluding exon 20 insertions) and accounting for compound mutations. *Classical* includes exon 19 deletions, as well as L858R and all T790M-containing mutations. *PACC* includes single and compound mutations, including PACC + classical. *Other uncommon* includes classical-like mutations (eg, L861Q), transmembrane, extracellular, and other mutations.

Methods

- The AACR Project GENIE v13 dataset (The AACR Project GENIE Consortium, 2017) was used for mutation analysis in NSCLC patients. The database contains mutation data for patients tested across multiple academic sites in the United States, Canada, and Europe. At the time of the analysis, GENIE v13 contained Next Generation Sequencing (NGS) results for 19,529 NSCLC patients and represented a mixed population of treatment-naïve and pretreated patients.
- Firmonertinib in vitro inhibitory activity was tested by cell viability assay in Ba/F3 engineered cell lines expressing EGFR mutations at MD Anderson Cancer Center.
- The in vivo anti-tumor activity of firmonertinib was assessed in patient-derived xenograft (PDX) models of EGFR exon 20 insertion and PACC mutations (WuXi AppTec).
- Molecular modeling of firmonertinib and osimertinib in EGFR D770_N771insNPG (exon 20 insertion) and G719S (PACC) mutants was performed with Molecular Operating Environment (MOE) software (Chemical Computing Group) using 4LRM and 2ITN EGFR crystal structures obtained from Protein Data Bank (PDB).

Results

Figure 2. EGFR exon 20 insertion mutations represent a diverse group of insertion mutations

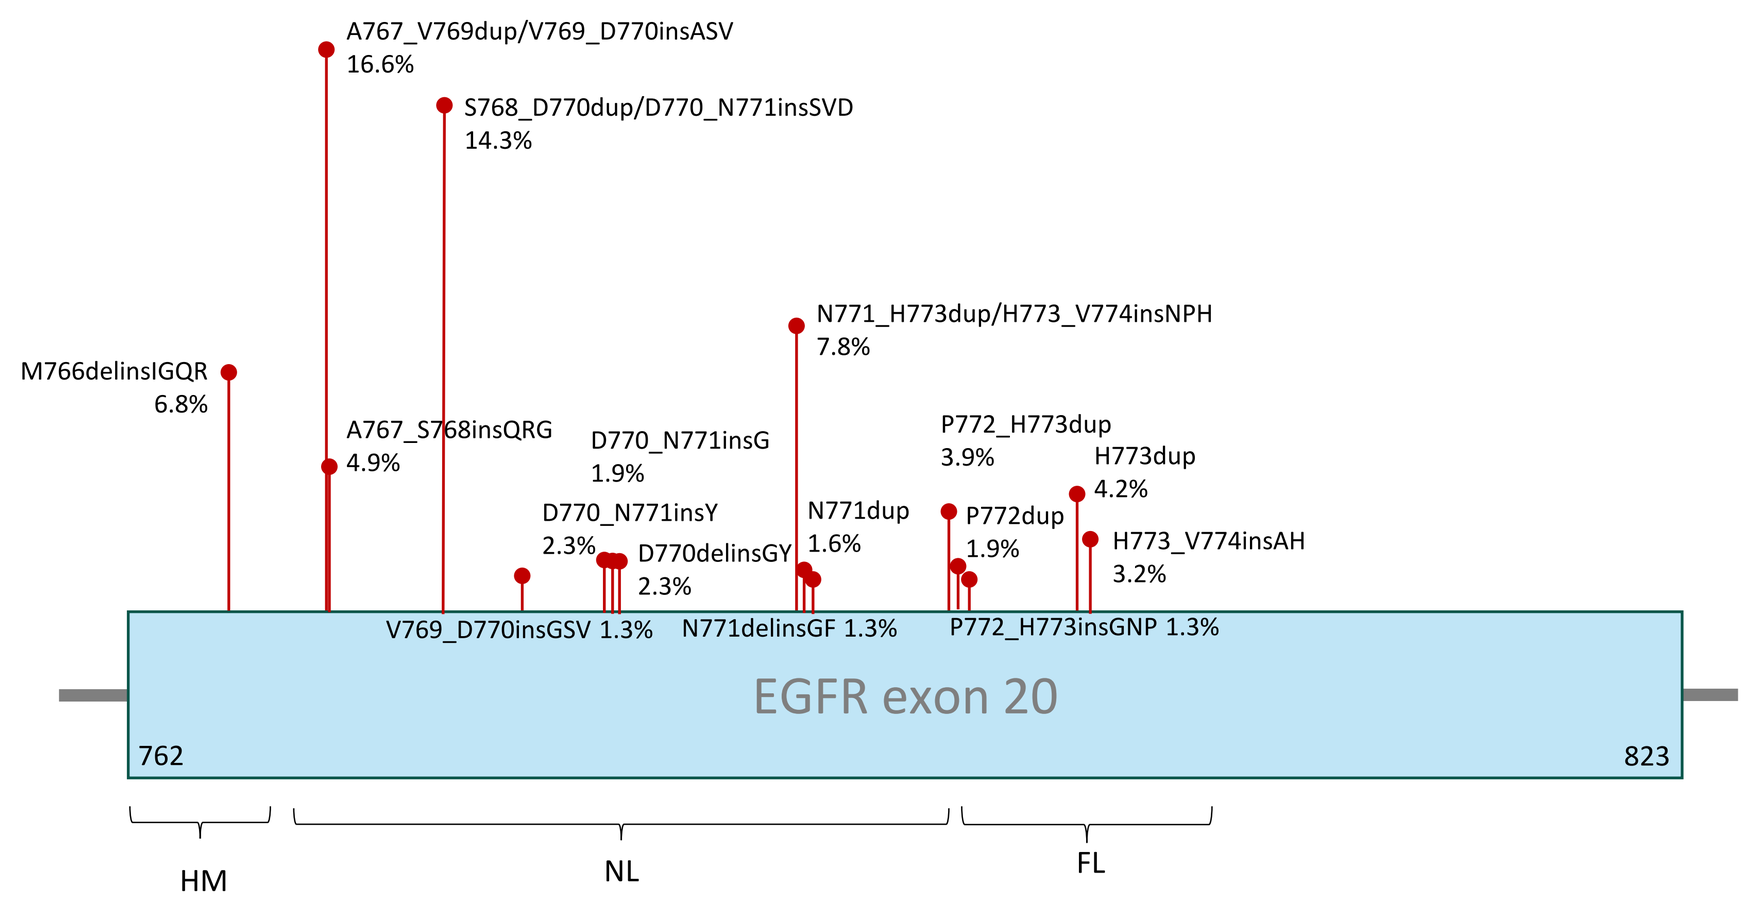
A767_V769dup/V769_D770insASV 16.6%
S768_D770dup/D770_N771insSVD 14.3%
M766delinsIGQR 6.8%
A767_S768insQRG 4.9%
N771_H773dup/H773_V774insNPH 7.8%
D770_N771insG 1.9%
D770_N771insY 2.3%
D770delinsGY 2.3%
N771dup 1.6%
P772_H773dup 3.9%
P772dup 1.9%
H773dup 4.2%
H773_V774insAH 3.2%
V769_D770insGSV 1.3%
N771delinsGF 1.3%
P772_H773insGNP 1.3%
EGFR exon 20
762
823
HM
NL
FL

Frequency of the most common (> 1%) EGFR exon 20 insertion mutations among all exon 20 insertion mutations in NSCLC found in the GENIE dataset (N = 307). HM, NL, and FL indicate regions with helical, near loop, and far loop exon 20 insertion mutations. The analysis was conducted based on EGFR exon 20 insertion mutations identified in patients with NSCLC in GENIE dataset v13.

Figure 3. EGFR PACC mutations represent a diverse group of missense mutations

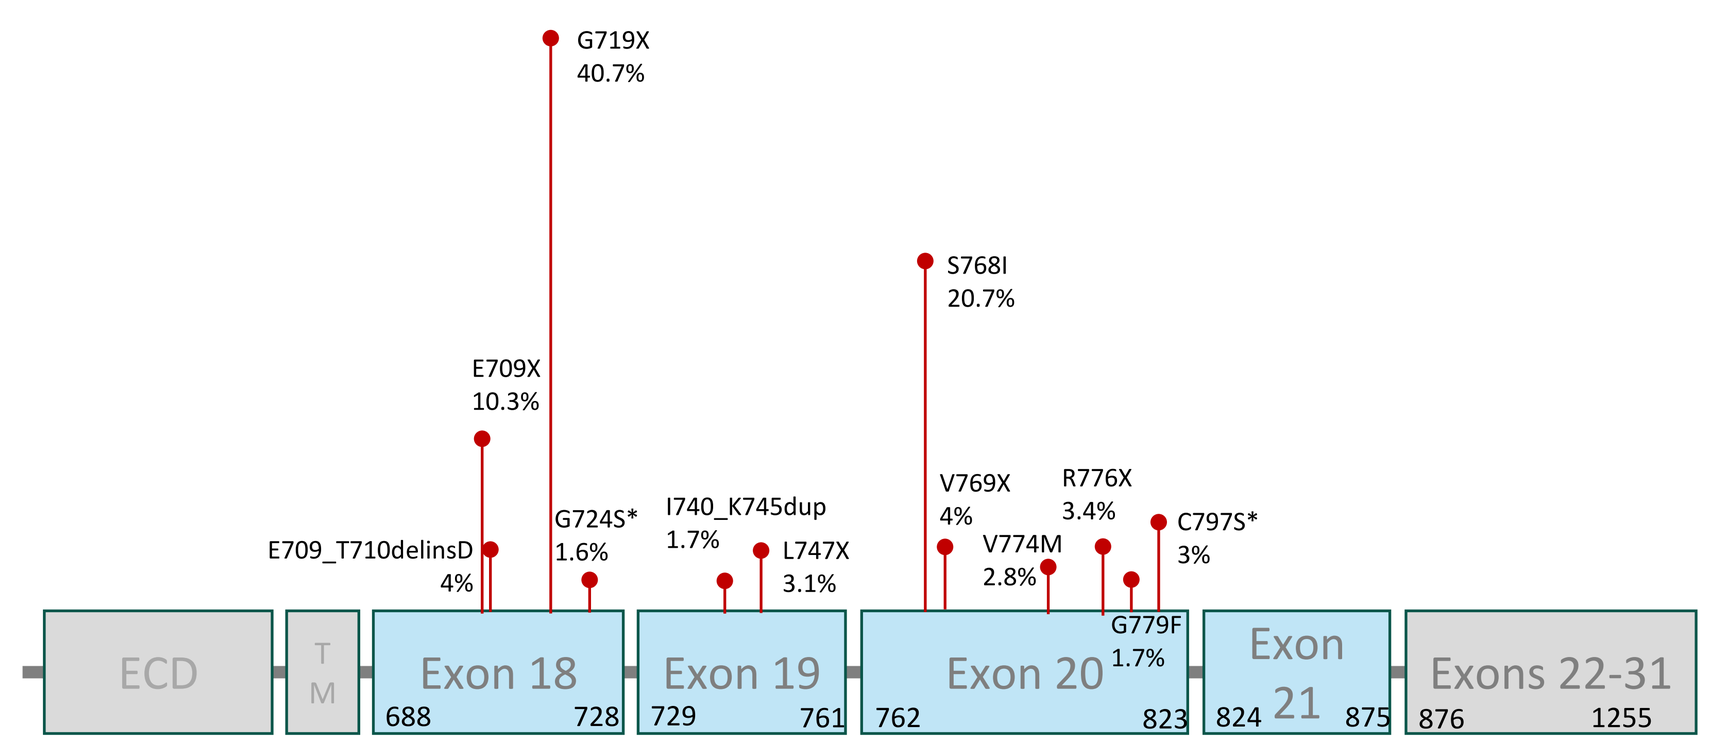
G719X 40.7%
S768I 20.7%
E709X 10.3%
E709_T710delinsD 4%
G724S* 1.6%
I740_K745dup 1.7%
L747X 3.1%
V769X 4%
V774M 2.8%
R776X 3.4%
C797S* 3%
G779F 1.7%
ECD
TM
Exon 18
Exon 19
Exon 20
Exon 21
Exons 22-31
688
728
729
761
762
823
824
875
876
1255

*G724S and C797S mutations have been reported as resistance mutations to osimertinib.

Frequency of the most common (> 1%) EGFR PACC mutations among all PACC mutations in NSCLC found in the GENIE dataset (N = 643). X, any amino acid; ECD, extracellular domain; TM, transmembrane region. The analysis was conducted based on PACC mutations identified in GENIE dataset v13 in patients with NSCLC.

Figure 4. EGFR PACC mutations are found as single or compound mutations in NSCLC

PACC Mutation Subtype	Examples of PACC mutation variants
PACC alone	G719X S768I E709X E709_T710delinsD G724S L747X K757X V774M I740_K745dup/I740dupIPVAIK V765L V769X R776X G779F
PACC + PACC	G719X + S768I S768I + V769L S768I + G724S
PACC + classical/classical-like	S768I + L861Q E709A + L858R R776H + L861Q
PACC + other EGFR mutation	V774M + H773L S768I + S784Y

Percent of NSCLC patients with each PACC subtype

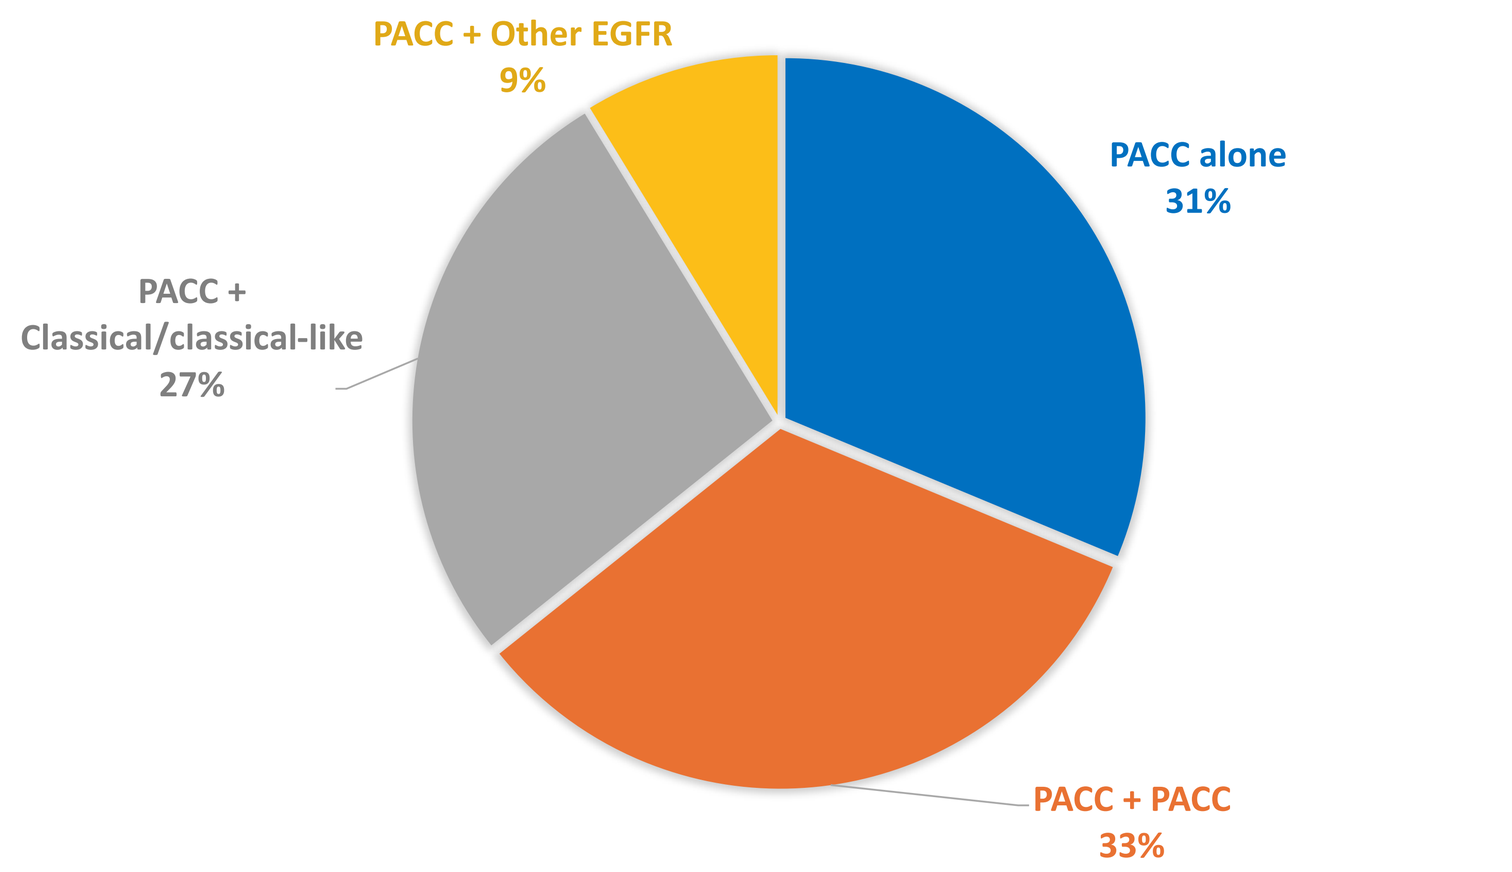
PACC + Other EGFR
9%
PACC alone
31%
PACC +
Classical/classical-like
27%
PACC + PACC
33%

Classical/classical-like includes classical (exon 19 deletions, L858R) and classical-like (L861Q L833V, T725M, and others) mutations.
Other includes other EGFR mutations, including those in the extracellular and transmembrane domain, as well as in exons 22–31.
Analysis based on PACC mutation data from AACR GENIE v13 in patients with NSCLC (N = 470).

Figure 5. Genetic co-alterations with EGFR PACC mutations are similar to co-alterations with exon 20 insertions and classical/classical-like EGFR mutations

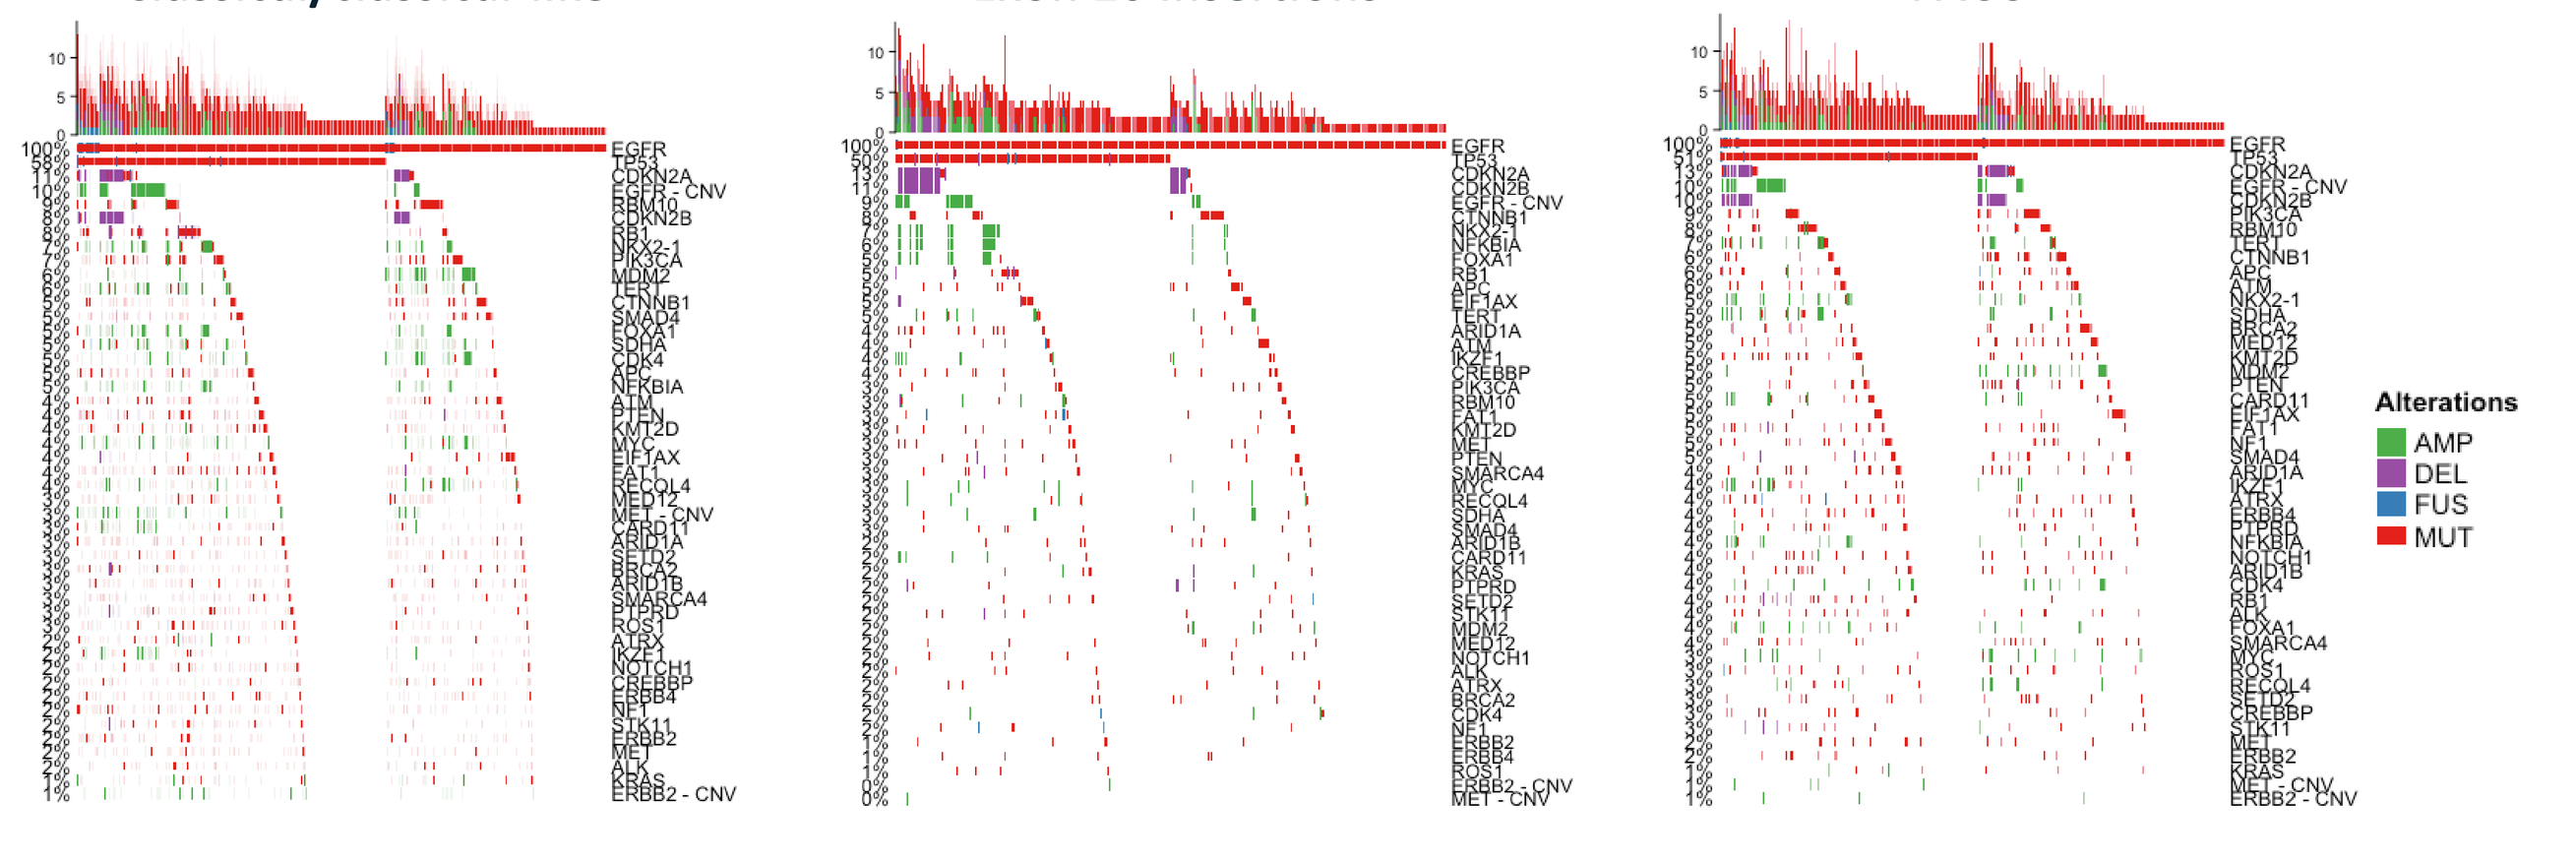
Classical/classical-like
Exon 20 insertions
PACC
Alterations
AMP
DEL
FUS
MUT

The genes shown in the plots are the most frequently co-mutated genes with EGFR mutations in NSCLC tumors. Alterations include single nucleotide variants (SNVs), deletion/insertions and splice variants (labelled as MUT), and fusions (FUS), as well as copy number variations (CNVs): amplifications (AMPs) and deletions (DELs). *Classical/classical-like* includes classical (exon 19 deletions and L858R) and classical-like (L861Q, L833V, T725M, and other) mutations. *PACC* mutations include single and compound mutations including those with classical/classical-like EGFR mutations. Co-mutation analysis was performed using mutation data from GENIE v13.

Figure 6. Firmonertinib is highly active against classical and uncommon EGFR mutations in vitro

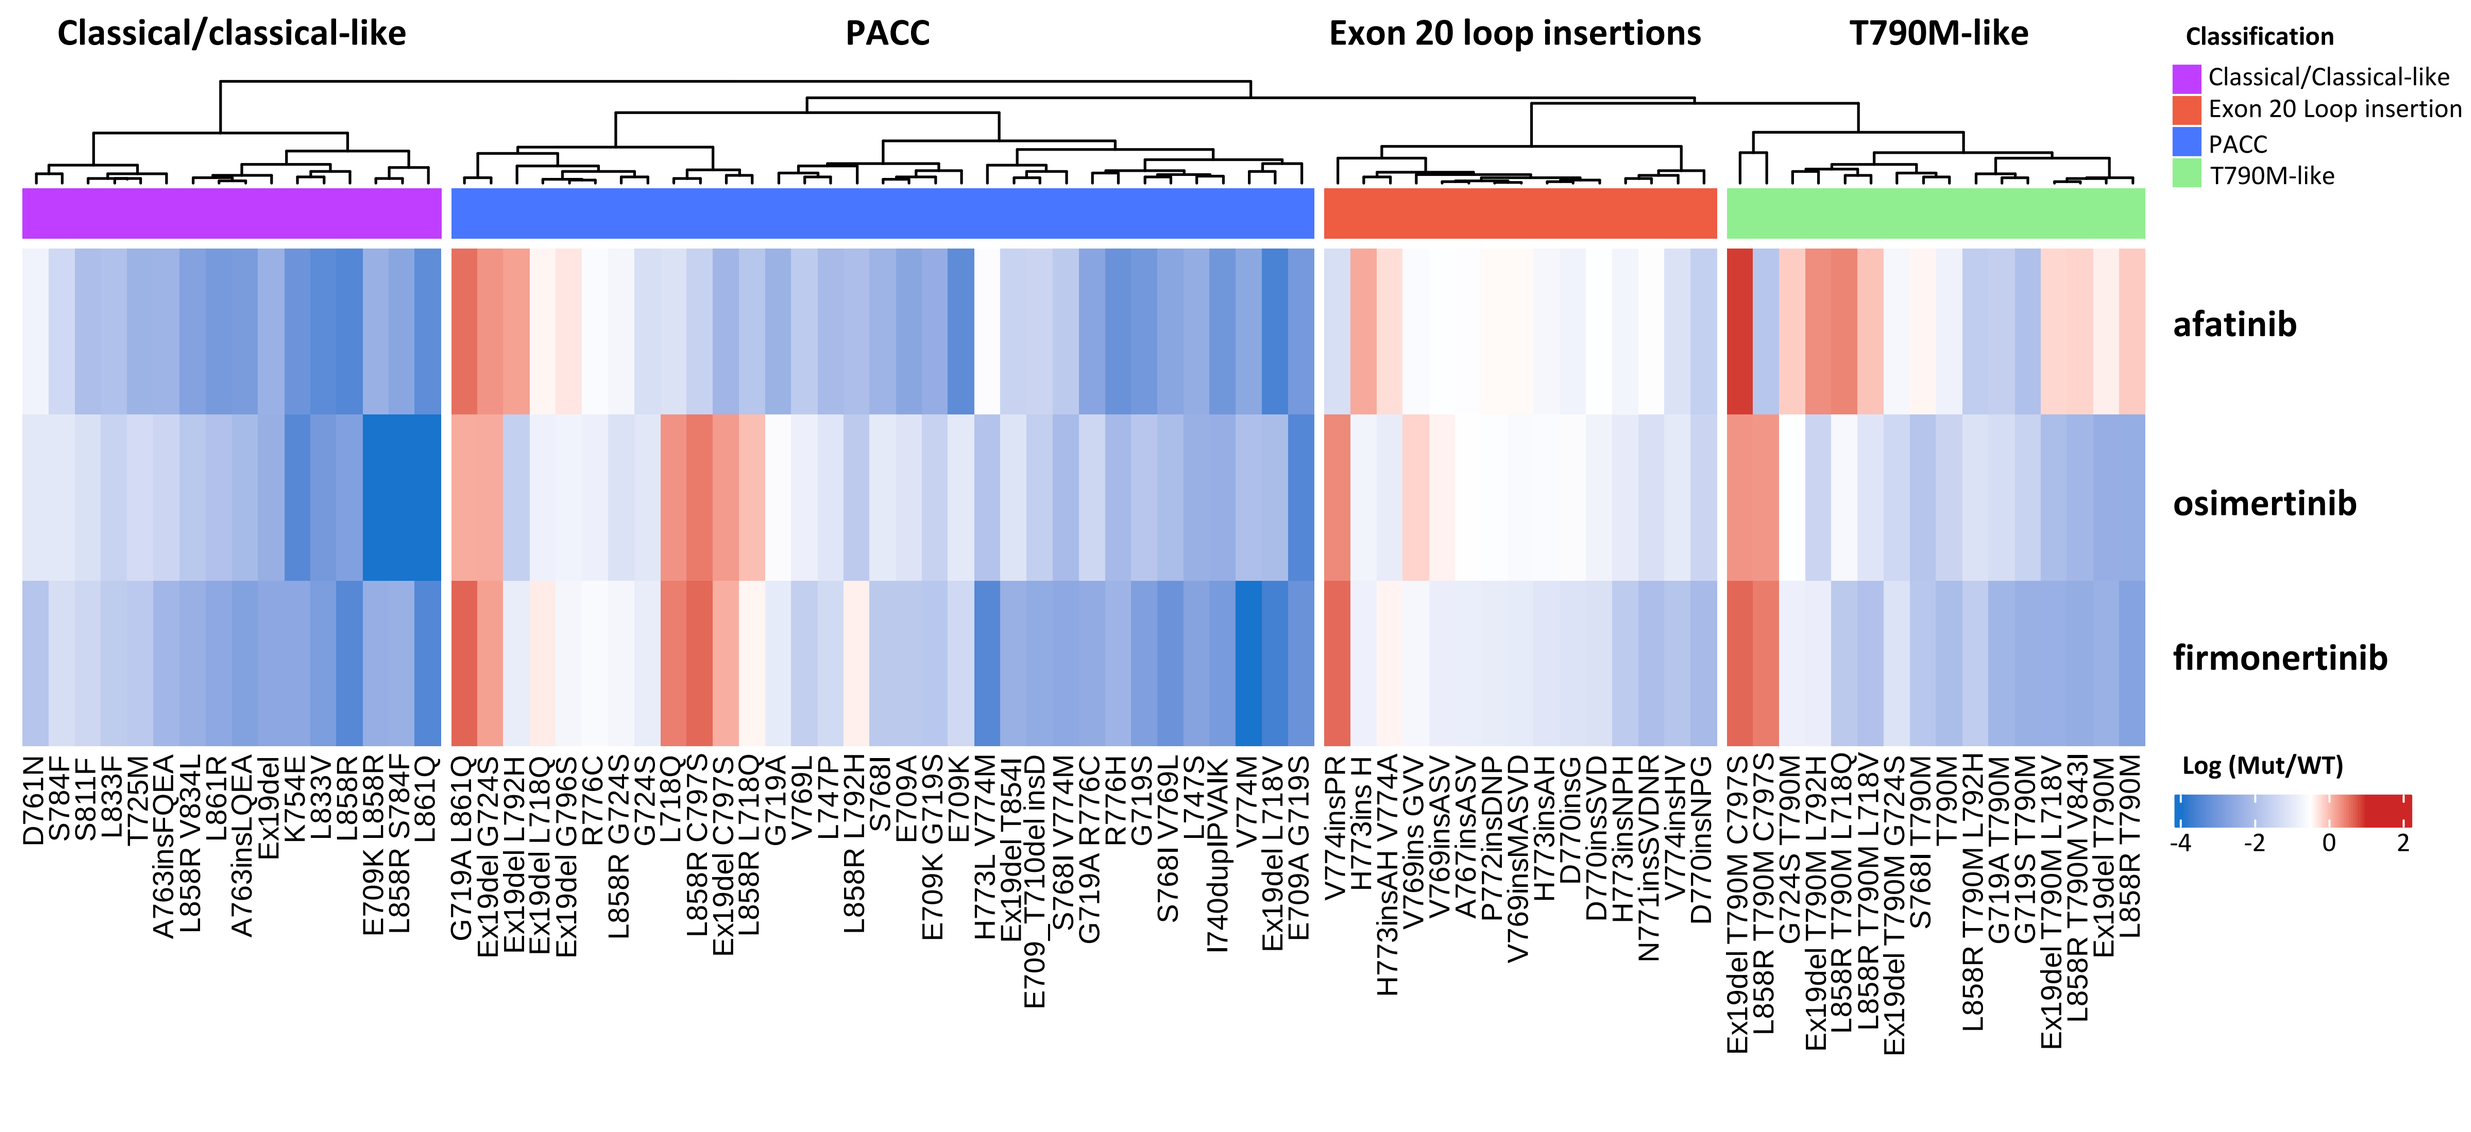
Classical/classical-like
PACC
Exon 20 loop insertions
T790M-like
Classification
Classical/Classical-like
Exon 20 Loop insertion
PACC
T790M-like
afatinib
osimertinib
firmonertinib
Log (Mut/WT)

- The activity of firmonertinib and other EGFR targeting agents (osimertinib and afatinib) was evaluated across various EGFR mutations and normalized to EGFR WT. The mutant/wild-type IC_{50} ratio is shown.
- For exon 20 loop insertions and PACC mutants, firmonertinib showed improved mutant/wild-type IC_{50} ratios as compared to osimertinib for most mutations.
- Firmonertinib also demonstrated promising mutant/wild-type IC_{50} ratios for classical/classical-like mutated EGFR.

Figure 7. Firmonertinib is highly active in vitro against the most prevalent EGFR exon 20 insertion mutations

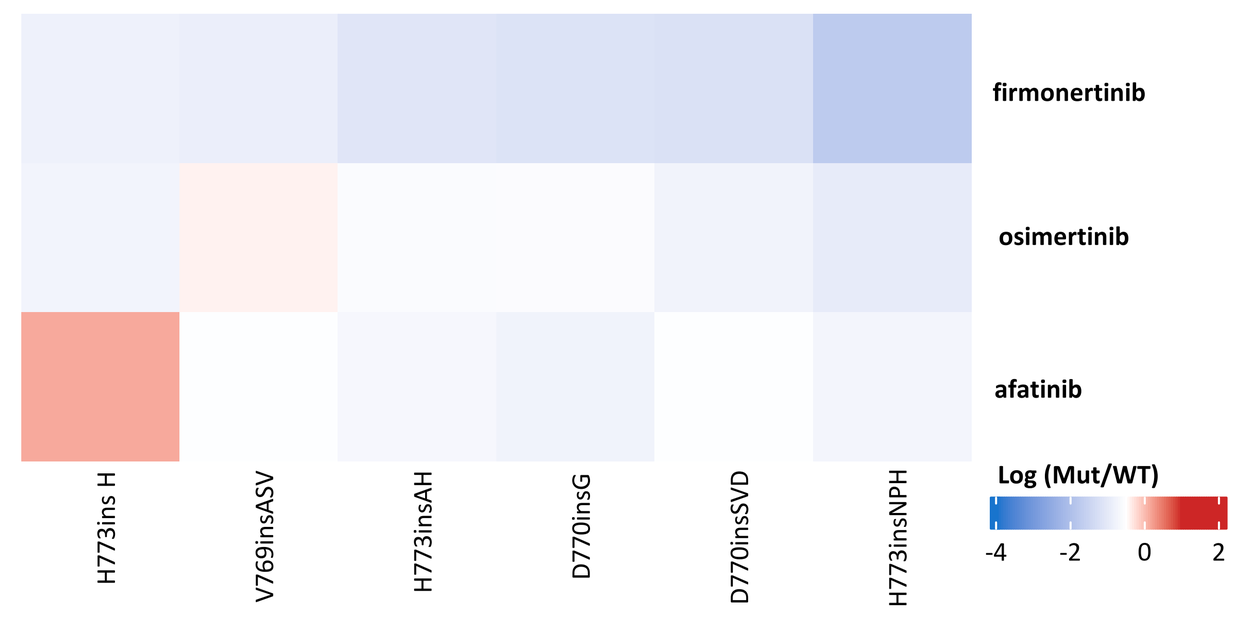
firmonertinib
osimertinib
afatinib
H773ins H
V769insASV
H773insAH
D770insG
D770insSVD
H773insNPH
Log (Mut/WT)

- Among the most prevalent exon 20 insertions found in patients (>1% mutation frequency), firmonertinib shows sensitivity that is superior to both osimertinib and afatinib across all mutations through the improved mutant/wild-type IC_{50} ratios.

Figure 8. Firmonertinib is highly active in vitro against the most prevalent EGFR PACC mutations

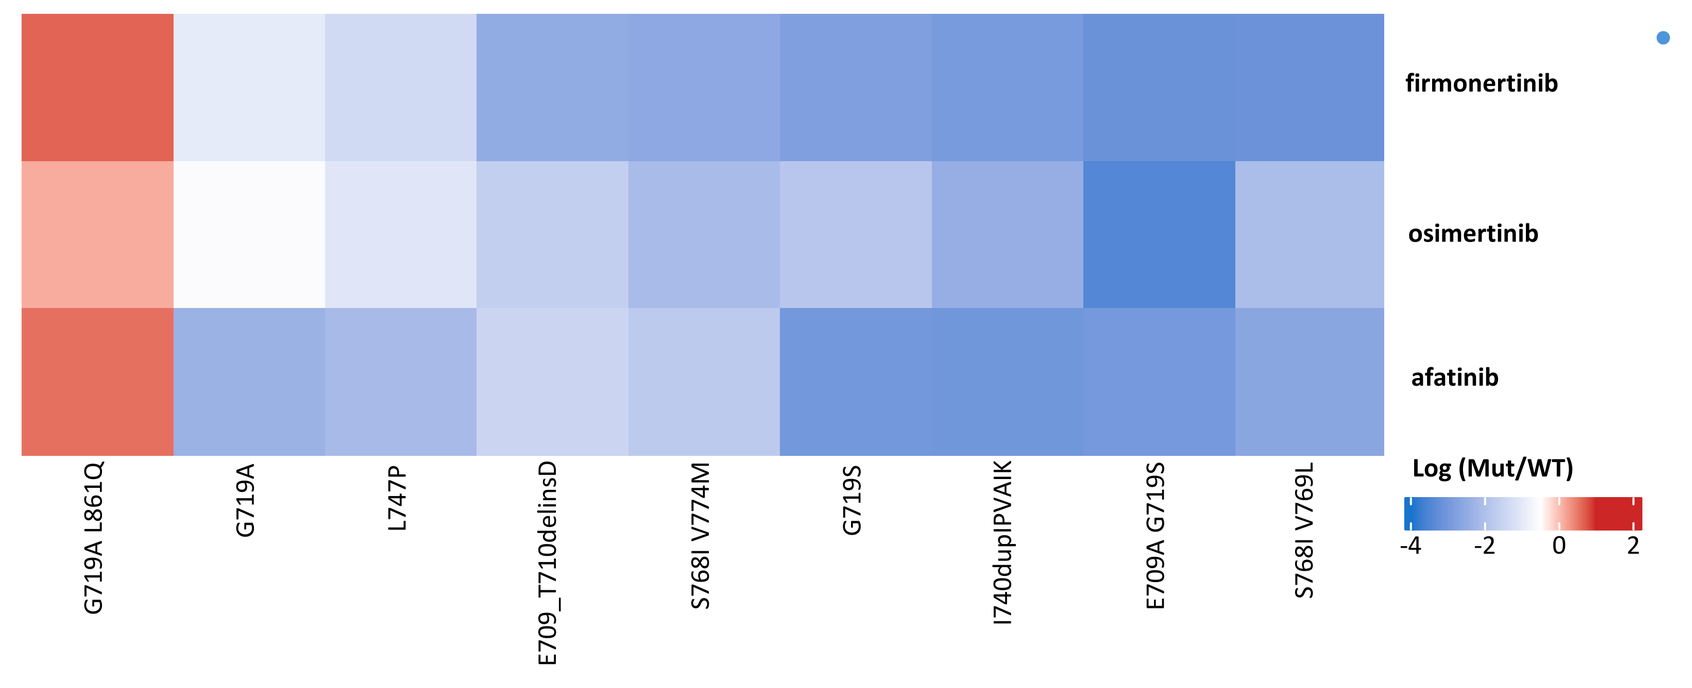
firmonertinib
osimertinib
afatinib
G719A L861Q
G719A
L747P
E709_T710delinsD
S768I V774M
G719S
I740dupIPVAIK
E709A G719S
S768I V769L
Log (Mut/WT)

- Among the most prevalent PACC mutations found in patients (>1% mutation frequency), firmonertinib shows sensitivity that is superior to osimertinib across all mutants (except for E709A + G719S) through the improved mutant/wild-type IC_{50} ratios and a similar sensitivity as afatinib. Note that G719A+L861Q was resistant in vitro to all inhibitors that were tested.

Table 1. Firmonertinib at clinically relevant drug levels inhibits PACC and exon 20 insertion mutations based on in vitro IC_{50} values

	Firmonertinib median IC_{50} [μM]*	Average $C_{min,ss}$ for firmonertinib [μM]		Average $C_{min,ss}$ for total analyte (firmonertinib + active metabolite) [μM]	
		160 mg clinical dose	240 mg clinical dose	160 mg clinical dose	240 mg clinical dose
Exon 20 insertions	0.0716	0.0860	0.0948	0.1711	0.2349
PACC	0.0022				

*median IC_{50} for the most prevalent mutations
$C_{min,ss}$ - minimum drug concentration at steady-state

- Average $C_{min,ss}$ for firmonertinib and for total analyte (firmonertinib and its active metabolite) cover the median IC_{50} values for the most prevalent exon 20 insertion and PACC mutant EGFR
- The minimum concentrations at steady-state were based on NSCLC patients enrolled in firmonertinib phase 1 and 1/2 studies, NCT02973763 and NCT03127449, respectively (Shi et al, 2020)

Figure 9. Firmonertinib is predicted to bind to a) exon 20 insertion and b) PACC mutated EGFR via additional contact points provided by the unique side chain

a) Exon 20 insertion

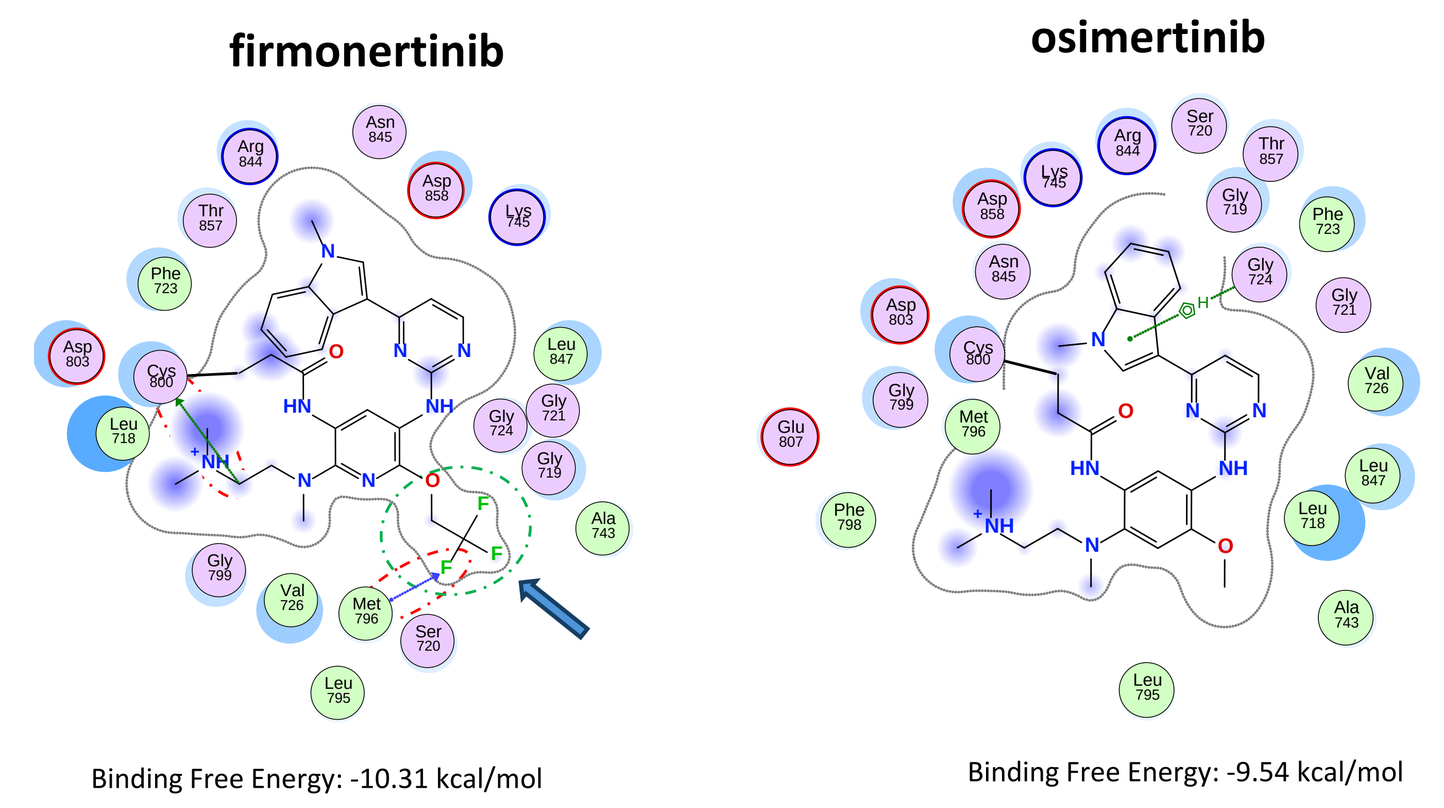
firmonertinib
osimertinib
Binding Free Energy: -10.31 kcal/mol
Binding Free Energy: -9.54 kcal/mol

- 2D molecular models of firmonertinib vs osimertinib bound to EGFR D770_N771insNPG mutant
- Red dotted outlines indicate hydrogen bonds between firmonertinib and EGFR (not present with osimertinib), which contribute to an improved interaction between firmonertinib and the binding pocket of EGFR.
- The proximity contour (green dotted outline) around firmonertinib's tri-fluoroethoxy side chain (blue arrow) confirms firmonertinib's close contact with the binding pocket of EGFR.

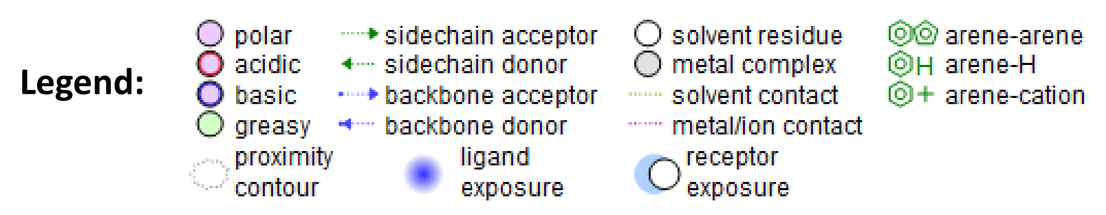
Legend:
polar
acidic
basic
greasy
proximity contour
sidechain acceptor
sidechain donor
backbone acceptor
backbone donor
ligand exposure
solvent residue
metal complex
solvent contact
metal/ion contact
receptor exposure
arene-arene
arene-H
arene-cation

b) PACC

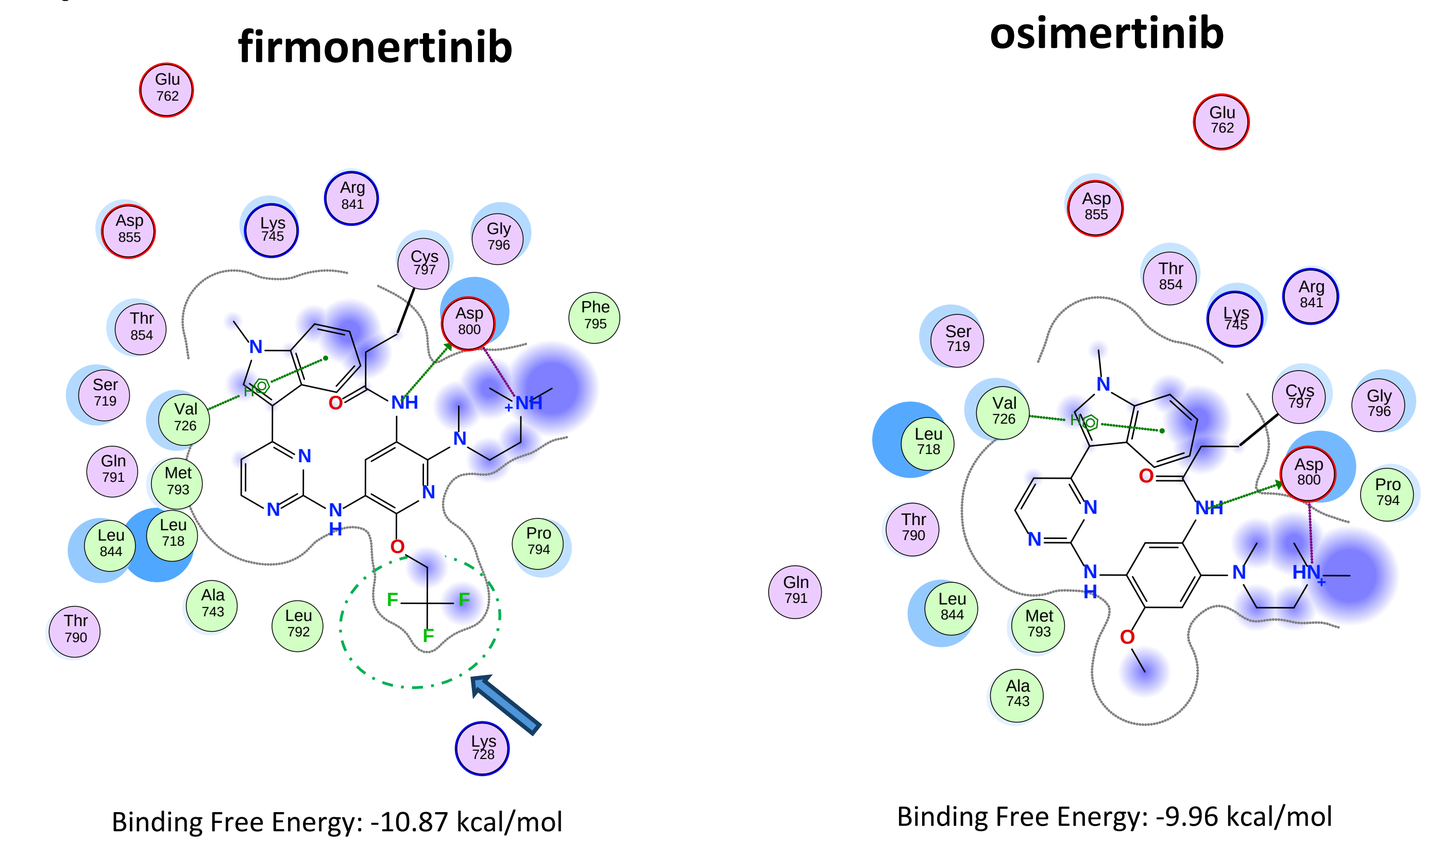
firmonertinib
osimertinib
Binding Free Energy: -10.87 kcal/mol
Binding Free Energy: -9.96 kcal/mol

- 2D molecular models of firmonertinib vs osimertinib bound to EGFR G719S mutant
- Green dotted outline indicates the proximity contour around the tri-fluoroethoxy group (blue arrow), confirming firmonertinib's close contact with the binding pocket and hydrophobic residues of EGFR (not present in osimertinib).
- Lower binding free energy for firmonertinib compared to osimertinib may correspond to the increase in the contact surface area between firmonertinib and EGFR.

Figure 10. Firmonertinib is superior to osimertinib against EGFR PACC mutations and exon 20 insertions in vitro

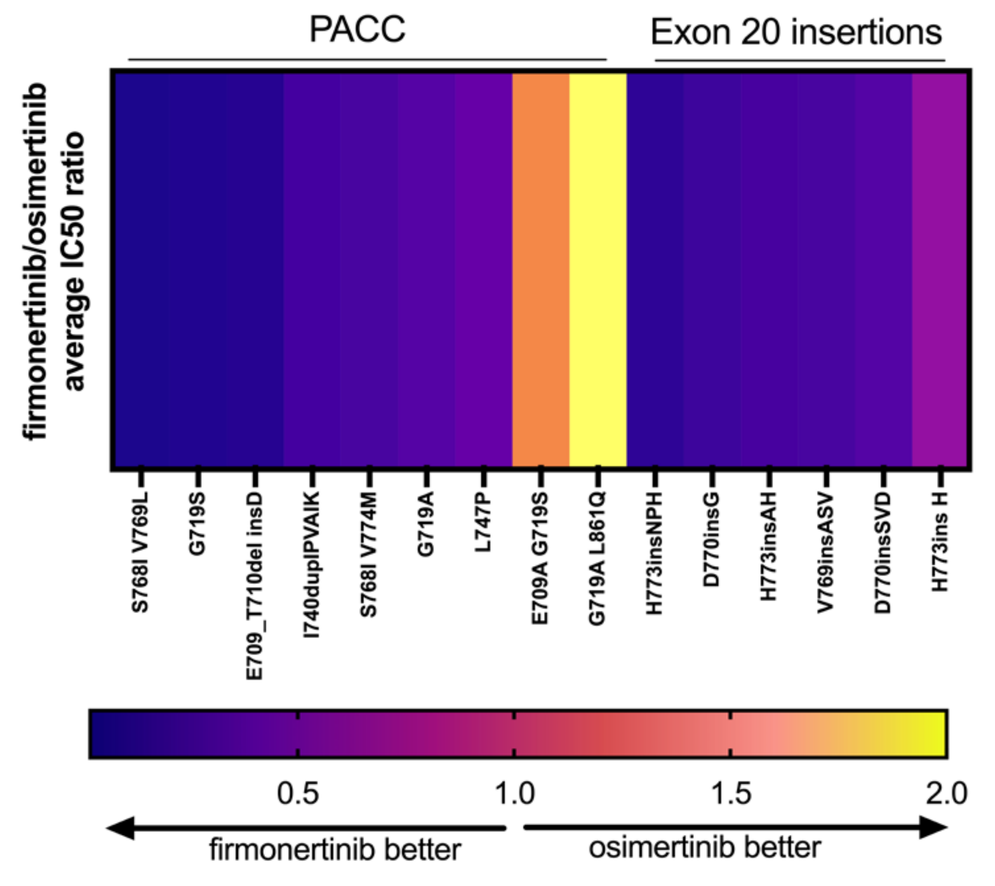
PACC
Exon 20 insertions
firmonertinib/osimertinib average IC50 ratio
S768I V769L
G719S
E709_T710del insD
I740dupIPVAIK
S768I V774M
G719A
L747P
E709A G719S
G719A L861Q
H773insNPH
D770insG
H773insAH
V769insASV
D770insSVD
H773ins H
0.5
1.0
1.5
2.0
firmonertinib better
osimertinib better

- The ratio of firmonertinib IC_{50} values to osimertinib IC_{50} values for the most prevalent PACC and exon 20 insertion mutations (>1%) demonstrate that firmonertinib is superior to osimertinib across all exon 20 insertion mutations (6 of 6) and in 78% of PACC mutations (7 of 9).
- Of note, the PACC-mutant G719A+L861Q was insensitive to both firmonertinib and osimertinib in these cell line models and had IC_{50} values above 1 μM.

Figure 11a. Firmonertinib is well tolerated and shows strong efficacy in a PACC PDX model harboring G719A + S768I compound mutation

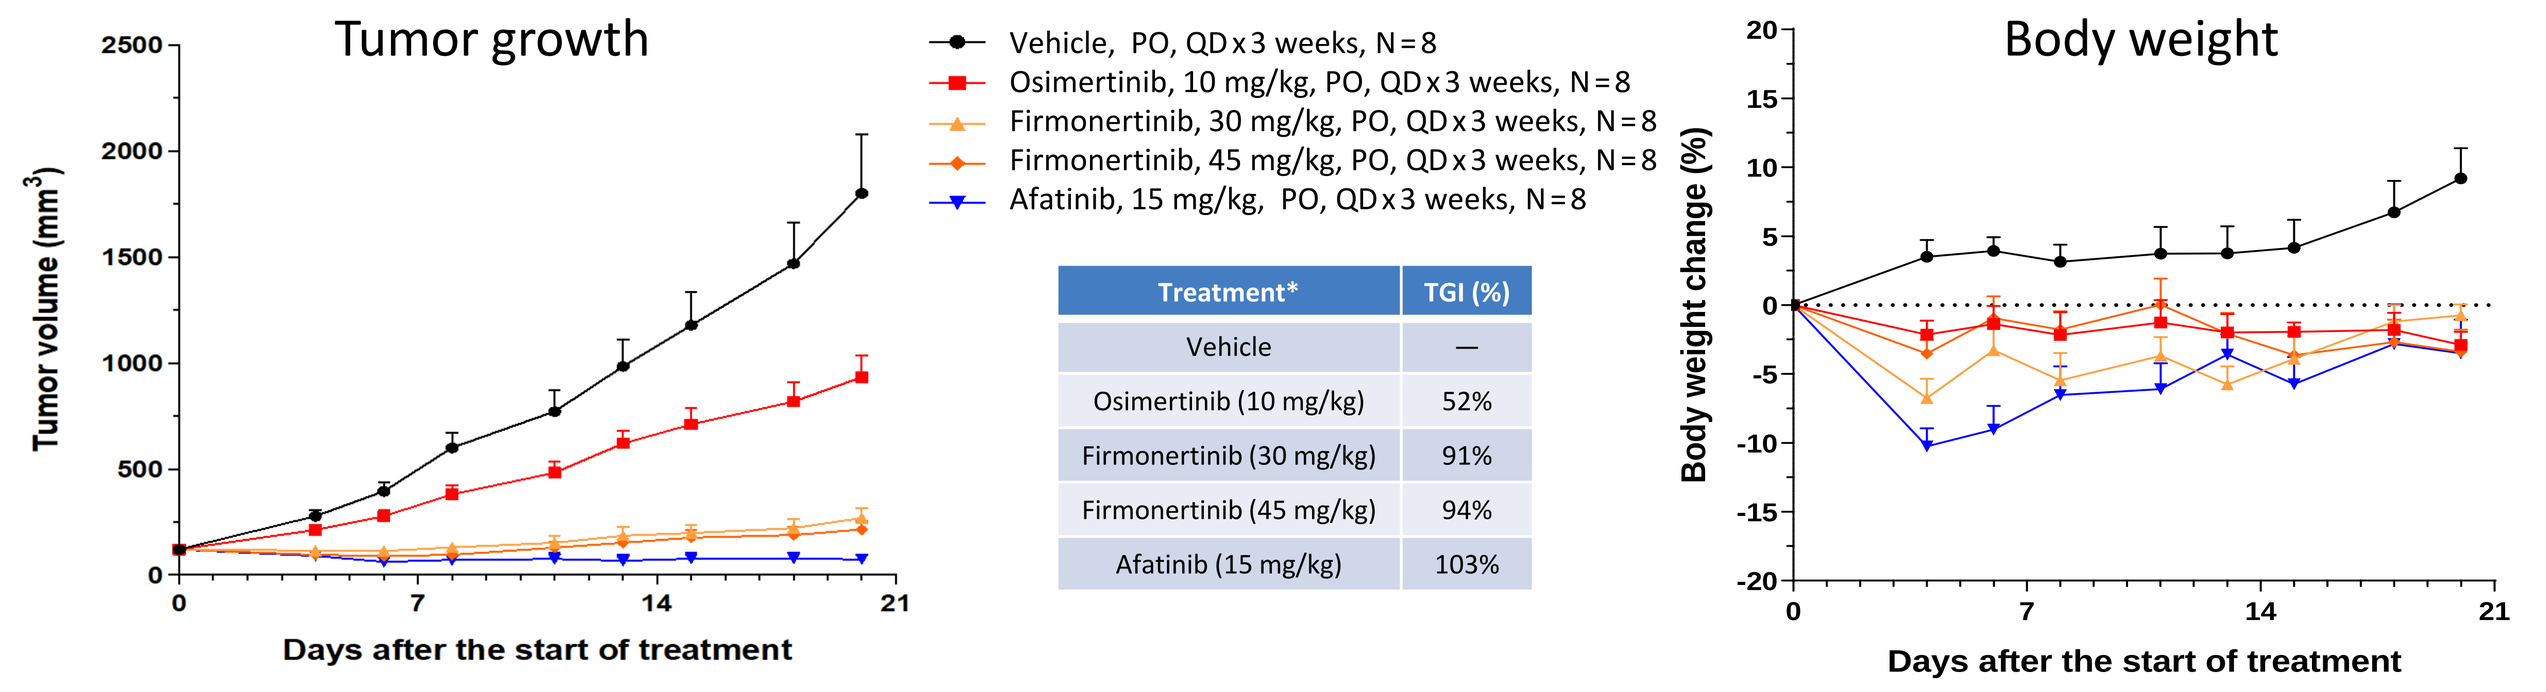
Tumor growth
Tumor volume (mm3)
Days after the start of treatment
Vehicle, PO, QD x 3 weeks, N=8
Osimertinib, 10 mg/kg, PO, QD x 3 weeks, N=8
Firmonertinib, 30 mg/kg, PO, QD x 3 weeks, N=8
Firmonertinib, 45 mg/kg, PO, QD x 3 weeks, N=8
Afatinib, 15 mg/kg, PO, QD x 3 weeks, N=8
Treatment* TGI (%)
Vehicle —
Osimertinib (10 mg/kg) 52%
Firmonertinib (30 mg/kg) 91%
Firmonertinib (45 mg/kg) 94%
Afatinib (15 mg/kg) 103%
Body weight
Body weight change (%)
Days after the start of treatment

- Firmonertinib treatment was well tolerated and significantly inhibited tumor growth in the G719A + S768I NSCLC PDX model up to 91% and 94% at the 30- and 45-mg/kg dose levels evaluated, respectively, while osimertinib showed minimal efficacy of 52% at a clinically relevant dose.
- Afatinib treatment had a slightly higher efficacy with tumor growth inhibition (TGI) of 103% in comparison with firmonertinib, but it was the least well tolerated treatment in this model.

Figure 11b. Firmonertinib is well tolerated and shows strong efficacy in a PACC + classical-like PDX model harboring a G719S + L861Q compound mutation

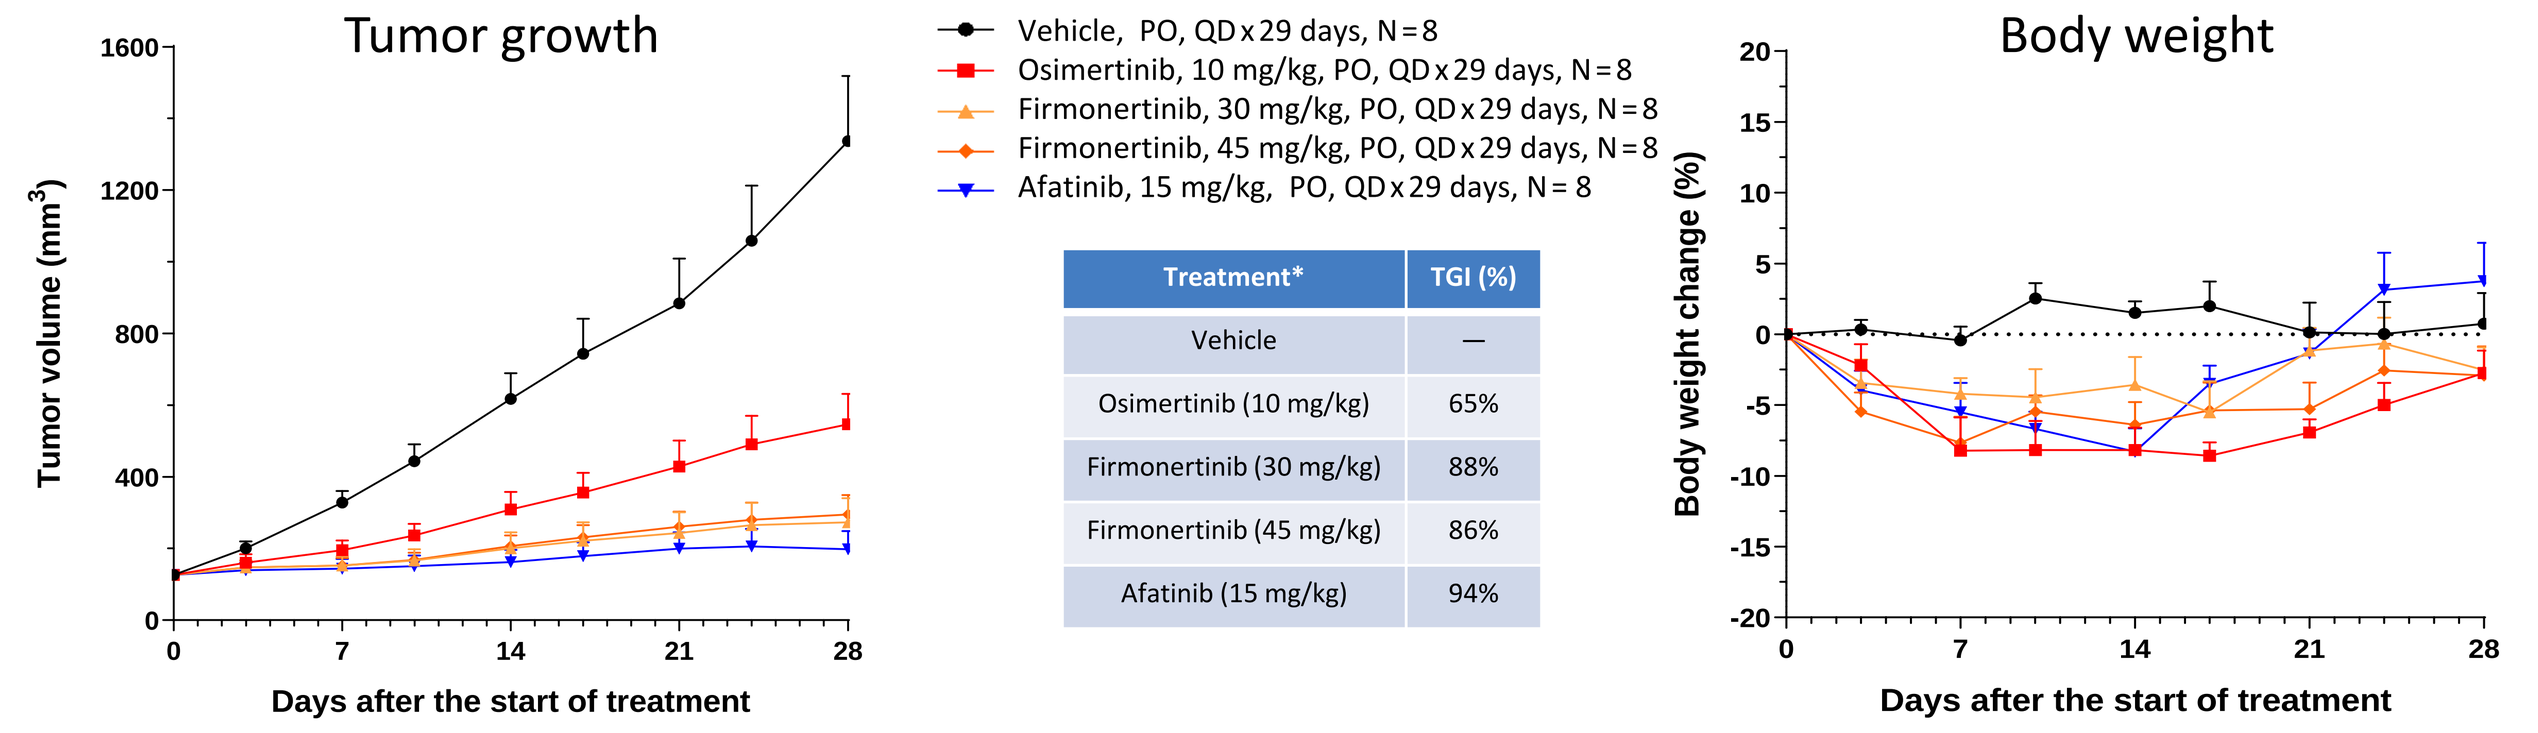
Tumor growth
Tumor volume (mm3)
Days after the start of treatment
Vehicle, PO, QD x 29 days, N=8
Osimertinib, 10 mg/kg, PO, QD x 29 days, N=8
Firmonertinib, 30 mg/kg, PO, QD x 29 days, N=8
Firmonertinib, 45 mg/kg, PO, QD x 29 days, N=8
Afatinib, 15 mg/kg, PO, QD x 29 days, N=8
Treatment* TGI (%)
Vehicle —
Osimertinib (10 mg/kg) 65%
Firmonertinib (30 mg/kg) 88%
Firmonertinib (45 mg/kg) 86%
Afatinib (15 mg/kg) 94%
Body weight
Body weight change (%)
Days after the start of treatment

- Firmonertinib treatment was generally well tolerated and significantly inhibited tumor growth in the G719S + L861Q NSCLC PDX model up to 88% and 86% at the 30- and 45-mg/kg dose levels evaluated, respectively, while osimertinib showed partial efficacy of 65% at a clinically relevant dose.
- Afatinib treatment had a slightly higher efficacy with tumor growth inhibition (TGI) of 94% in comparison with firmonertinib in this model.

*Firmonertinib doses of 30 and 45 mg/kg in mice are equivalent to approximately 160- and 240-mg clinical doses in humans, respectively, based on exposure (AUC; data on file). The osimertinib dose of 10 mg/kg and afatinib dose of 15 mg/kg were chosen as clinically relevant based on previous reports (Jänne et al, 2015; Ballard et al, 2016; Colclough et al, 2021; Wind et al, 2017).

Conclusions

- PACC mutations are important EGFR driver mutations that are more prevalent than exon 20 insertion mutations in NSCLC.
- Firmonertinib is similarly active in nonclinical models against both PACC and exon 20 insertion mutations.
- Based on molecular modeling, firmonertinib has increased contact points within the drug binding pocket of PACC and exon 20 insertion–mutant EGFR, consistent with the increased sensitivity observed in vitro and in vivo versus osimertinib.
- Firmonertinib at clinically relevant drug levels is predicted to inhibit PACC and exon 20 insertion–mutant EGFR based on in vitro cell line IC_{50} values.
- In PDX in vivo studies, firmonertinib showed superior efficacy compared with osimertinib.
- The activity of firmonertinib in EGFR PACC–mutant NSCLC is currently being evaluated in the global study FURTHER (FURMO-002; NCT05364073). See abstract CT152, Section 50, Monday, April 8th, 1:30–5:00 PM.
- The activity of firmonertinib in EGFR exon 20 insertion–mutant NSCLC is currently being evaluated in the global randomized Phase 3 study FURVENT (FURMO-004; NCT05607550). See abstract CT280, Poster Session Section 50, Tuesday, April 9th, 1:30–5:00 PM.

FURTHER FURVENT

References

- Ballard et al. Preclinical comparison of osimertinib with other EGFR-TKIs in EGFR-mutant NSCLC brain metastases models, and early evidence of clinical brain metastases activity. *Clin Cancer Res*. 2016;22(20):5130–40.
- Friedlaender et al. EGFR and HER2 exon 20 insertions in solid tumours: from biology to treatment. *Nature Rev Clin Onc.* 2022; 19(1):51–69.
- Han et al. FAVOUR: A Phase 1b study of furmonertinib, an oral, brain penetrant, selective EGFR inhibitor, in patients with advanced NSCLC with EGFR exon 20 insertions. *J Thoracic Oncol.* 2023;18(11):s49
- Jänne et al. AZD9291 in EGFR inhibitor–resistant non-small-cell lung cancer. *N Engl J Med*. 2015;372:1689–99.
- Musib et al. PP01.11 Furmonertinib is an oral, irreversible, highly brain-penetrant pan-EGFR inhibitor with activity against classical and atypical EGFR mutations. *J Thoracic Oncol*. 2023;18(3):e14–e15.
- Robichaux et al. Structure-based classification predicts drug response in EGFR-mutant NSCLC. *Nature*. 2021;597(7878):732–7.
- The AACR Project GENIE Consortium. AACR Project GENIE: Powering precision medicine through an international consortium. *Cancer Discov.* 2017;7(8):818–31 (database version 13).
- Colclough et al. Preclinical Comparison of the Blood–brain barrier Permeability of Osimertinib with Other EGFR TKIs. *Clin Cancer Res.* 2021;27:189–201
- Wind et al. Clinical Pharmacokinetics and Pharmacodynamics of Afatinib. *Clin Pharmacokinet.* (2017) 56:235–250
- Shi et al. Safety, Clinical Activity, and Pharmacokinetics of Alflutinib (AST2818) in Patients With Advanced NSCLC With EGFR T790M Mutation. *J Thorac Oncol.* 2020 Jun;15(6):1015-1026.

Acknowledgments

- The authors acknowledge the American Association for Cancer Research and its financial and material support in the development of the AACR Project GENIE registry, as well as members of the Consortium for their commitment to data sharing. Interpretations are the responsibility of study authors.
- We thank J Ireland for supporting the mutation analysis of the GENIE database and James T. Metz for supporting the molecular modeling.

[1]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]ArriVent Biopharma Inc., Burlingame, CA, USA; [3]Allist Pharmaceuticals Co., Ltd., Shanghai, China